IDE161, a potential first-in-class clinical candidate PARG inhibitor, selectively targets homologous-recombination-deficient and PARP inhibitor resistant breast and ovarian tumors

Monah Abed, Diana Muñoz, Vidya Seshadri, Arjun A. Rao, Reeja Maskey, Steve Federowicz, Deepthi Bhupathi, Marya Liimatta, Rita Ousterhout, Firoz Jaipuri, Richard Zang, Christian Frey, Paul Barsanti, Claire Neilan, Mark Lackner, Mike White, Zineb Mounir

IDEAYA Biosciences, South San Francisco, CA USA

AACR Annual Meeting 2023, Poster 6093

Introduction

IDE161 is a potent, selective small molecule inhibitor of Poly (ADP-Ribose) Glycohydrolase (PARG) that is being developed as an anti-cancer therapeutic for patients with advanced or metastatic cancer harboring BRCA1/2 loss of function alterations and/or defects in homologous recombination repair pathway.

PARG plays an important role during the DNA damage repair process through hydrolysis of poly(ADP-ribose)(PAR) chains and accounts for approximately 90% of dePARylation activity within the cell. PAR chain synthesis is induced by poly(ADP-ribose) polymerases (PARPs) in response to DNA damage and replication stress and serves as a platform to recruit DNA repair proteins that are critical for PARP-mediated DNA repair processes. Following DNA repair, PARG hydrolyzes the poly(ADP-ribose) linkages to break down PAR chains, completing the cycle. Genetic deletion of PARG which is lethal during early embryogenesis as well as pharmacological inhibition of PARG lead to excessive PAR accumulation and subsequently to cell death in cells through delayed repair of DNA damage or incomplete DNA replication. This further highlights the importance of PARG during the process of DNA damage repair and supports the idea that PARG depletion or inhibition can exacerbate replication deficiencies making it a promising therapeutic target for a broad range of cancers with genomic instability.

Despite the overlapping pathway regulation driven by PARG and PARP, a differential sensitivity profile that relies on the balance of various fork stability components likely determines whether a cell is sensitive to PARP inhibitors, PARG inhibitors, or both. Thus, PARG inhibitors hold promise in targeting distinct tumor genotypes to PARP inhibitors in the clinic.

PARG activity resolves PARP-initiated DNA repair complexes to support stalled replication fork stability and restart

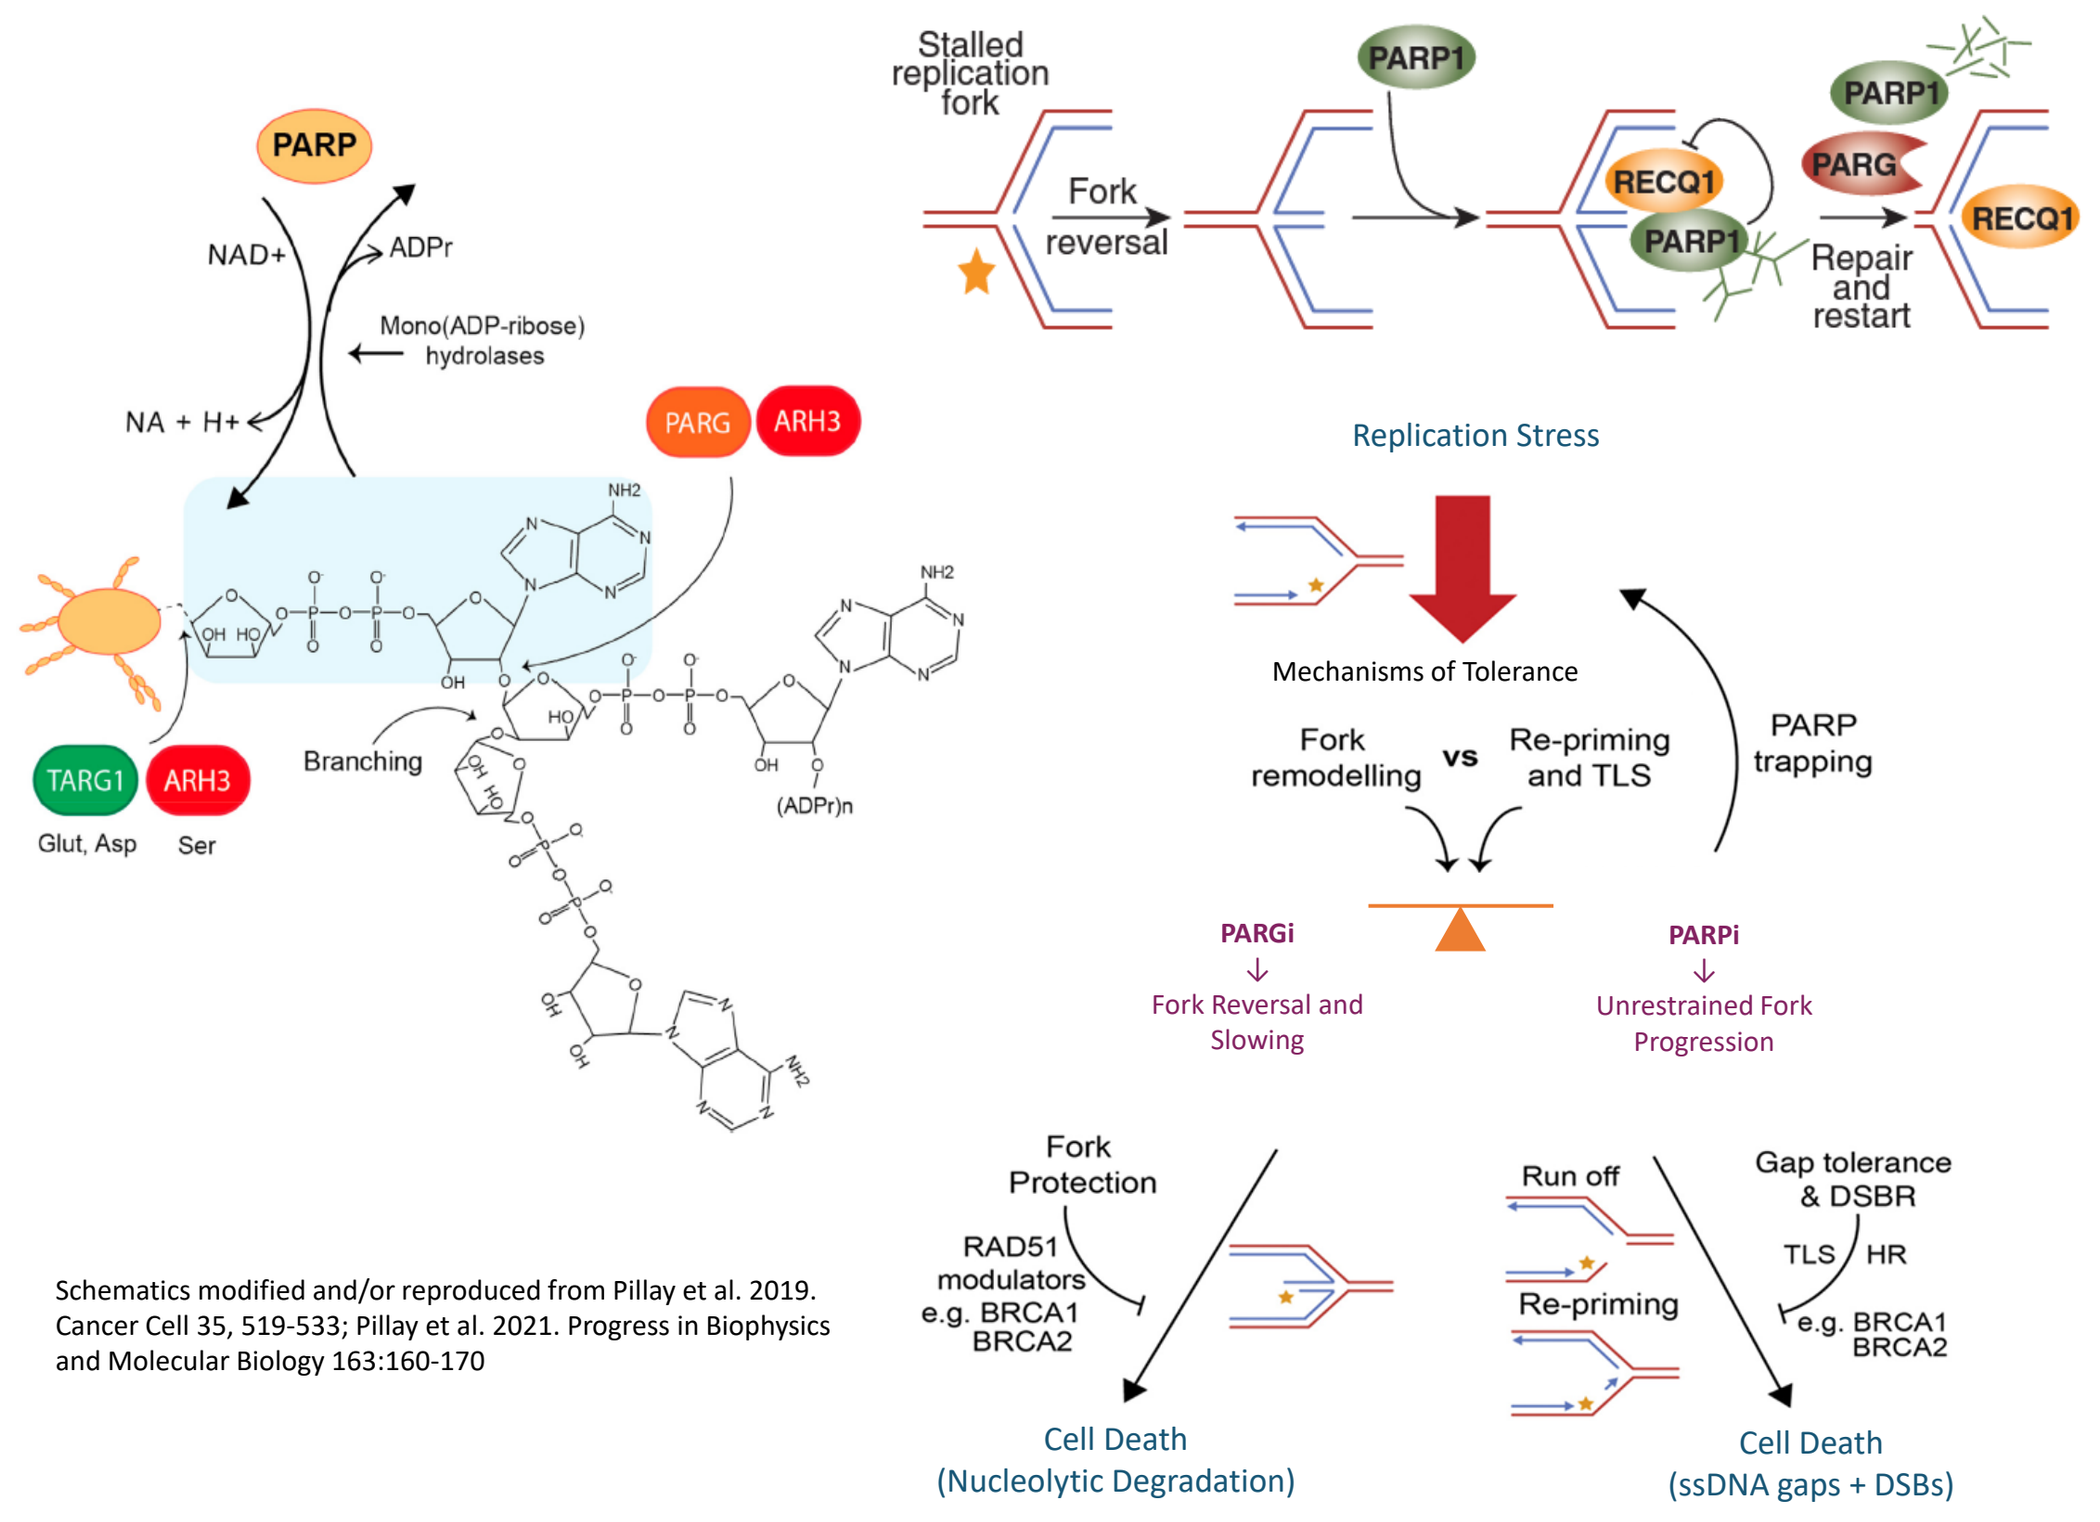
PARP
PARG
ARH3
TARG1
Stalled replication fork
Fork reversal
PARP1
RECQ1
Repair and restart
Replication Stress
Mechanisms of Tolerance
Fork remodelling
Re-priming and TLS
PARP trapping
PARGi
Fork Reversal and Slowing
PARPi
Unrestrained Fork Progression
Fork Protection
Cell Death (Nucleolytic Degradation)
Run off
Re-priming
Gap tolerance & DSBR
Cell Death (ssDNA gaps + DSBs)

Schematics modified and/or reproduced from Pillay et al. 2019. Cancer Cell 35, 519-533; Pillay et al. 2021. Progress in Biophysics and Molecular Biology 163:160-170

Summary

- Poly(ADP-Ribose) Glycohydrolase (PARG) plays a pivotal role in the regulation of DNA repair mechanisms acting in the same pathway as PARP and its inhibition can exacerbate replication deficiencies
- IDE161 is a potent and selective small molecule PARGi with anti-tumor activity in biomarker defined settings that are differentiated from PARP inhibitors. IDE161 shows activity in PARPi and platinum-resistant settings with a favorable non-clinical safety profile relative to PARPi
- ER+, Her2- breast cancer patients with HRD tumors are a strategic focus with unmet medical need (approximately 10% to 14% of breast cancer)
- Preclinical efficacy and tolerability supports a Ph 1 initial dose at one-half of the predicted human efficacious dose
- Clinical Ph 1/2 testing initiated: NCT05787587

References

- Cortes, U. et al. Mol Cell Biol. 2004; 24(16):7163-7178.
- Farmer, H. et al. Nature, 2005; 434(7035):917-921.
- Gao, H. et al. Exp Cell Res. 2007; 313(5):984-996.
- Gogola, E. et al. Cancer Cell 2018; 33(6):1078-1093.
- James, D. et al. ACS Chem Biol 2016; 11:3179–3190.
- McDermott, N. et al., Cancer Cell 2019; 35(3):344-346.
- Pillay, N. et al. Cancer Cell 2019; 35(3):519-533.
- Colic, M et al. Genome Medicine 2019; 11(52)
- Korotkevich G, et al. bioRxiv; DOI: 10.1101/060012.

Results

Figure 1: IDE161 is a potent, selective PARGi that demonstrates cellular activity in biomarker defined settings

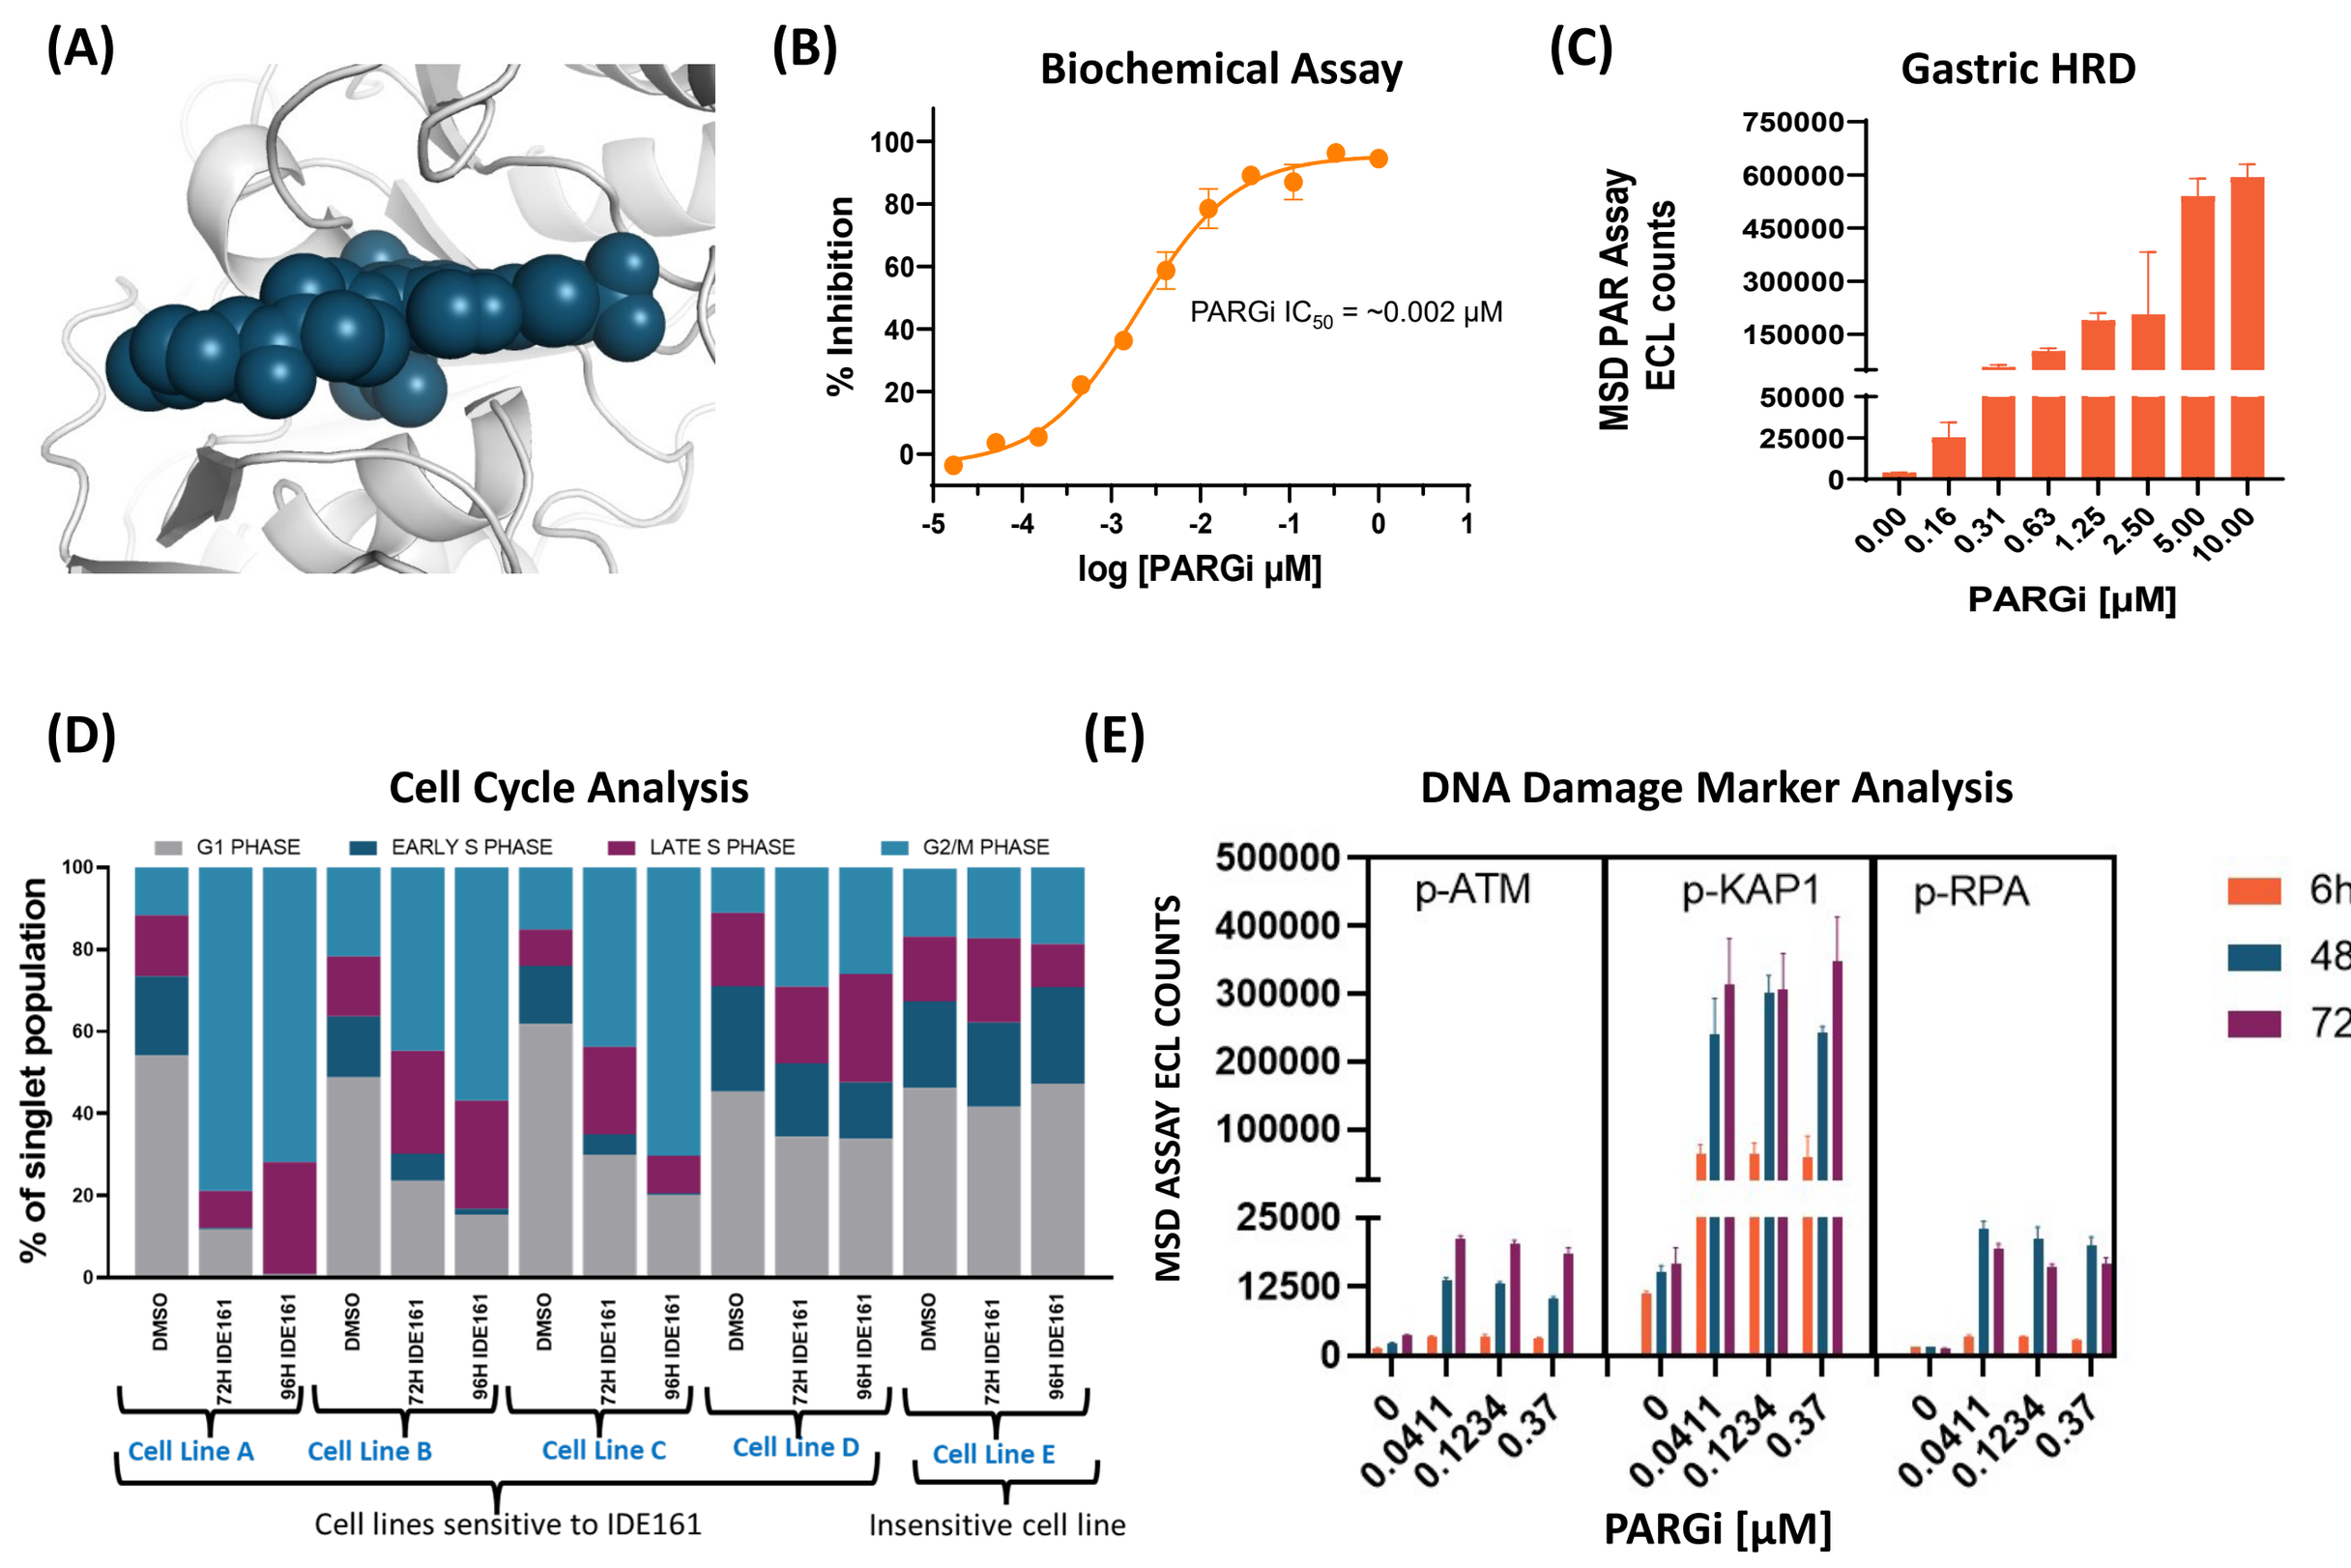
(A)
(B) Biochemical Assay
PARGi IC_{50} = ~0.002 µM
(C) Gastric HRD
(D) Cell Cycle Analysis
(E) DNA Damage Marker Analysis
p-ATM
p-KAP1
p-RPA
Cell lines sensitive to IDE161
Insensitive cell line

(A) Co-crystal structure of PARGi with PARG protein. (B) Determination of biochemical IC_{50} of IDE161 against human PARG showing IC_{50} of 2 nM. Data points are shown ± SEM. (C) PAR chain accumulation was evaluated using a PAR MSD assay in PARGi sensitive gastric HRD cell line. Values are plotted as mean ± SD. (D) Cell cycle effects of IDE161 were evaluated using flow cytometry in a panel of cell lines. (E) Accumulation of p-ATM, p-KAP1 and p-RPA was measured at indicated time points in PARGi sensitive cell lines using an MSD-based assay. Values are plotted as mean ± SD.

- IDE161 is a potent, selective small molecule PARG inhibitor
- IDE161 arrests cell cycle progression and induces DNA damage markers p-ATM, p-KAP1 and p-RPA consistent with DDR pathway activity
- Favorable ADME, physicochemical property and nonclinical-safety profile

Figure 2: IDE161 shows strong synthetic lethal interactions with key DNA repair genes across multiple indications

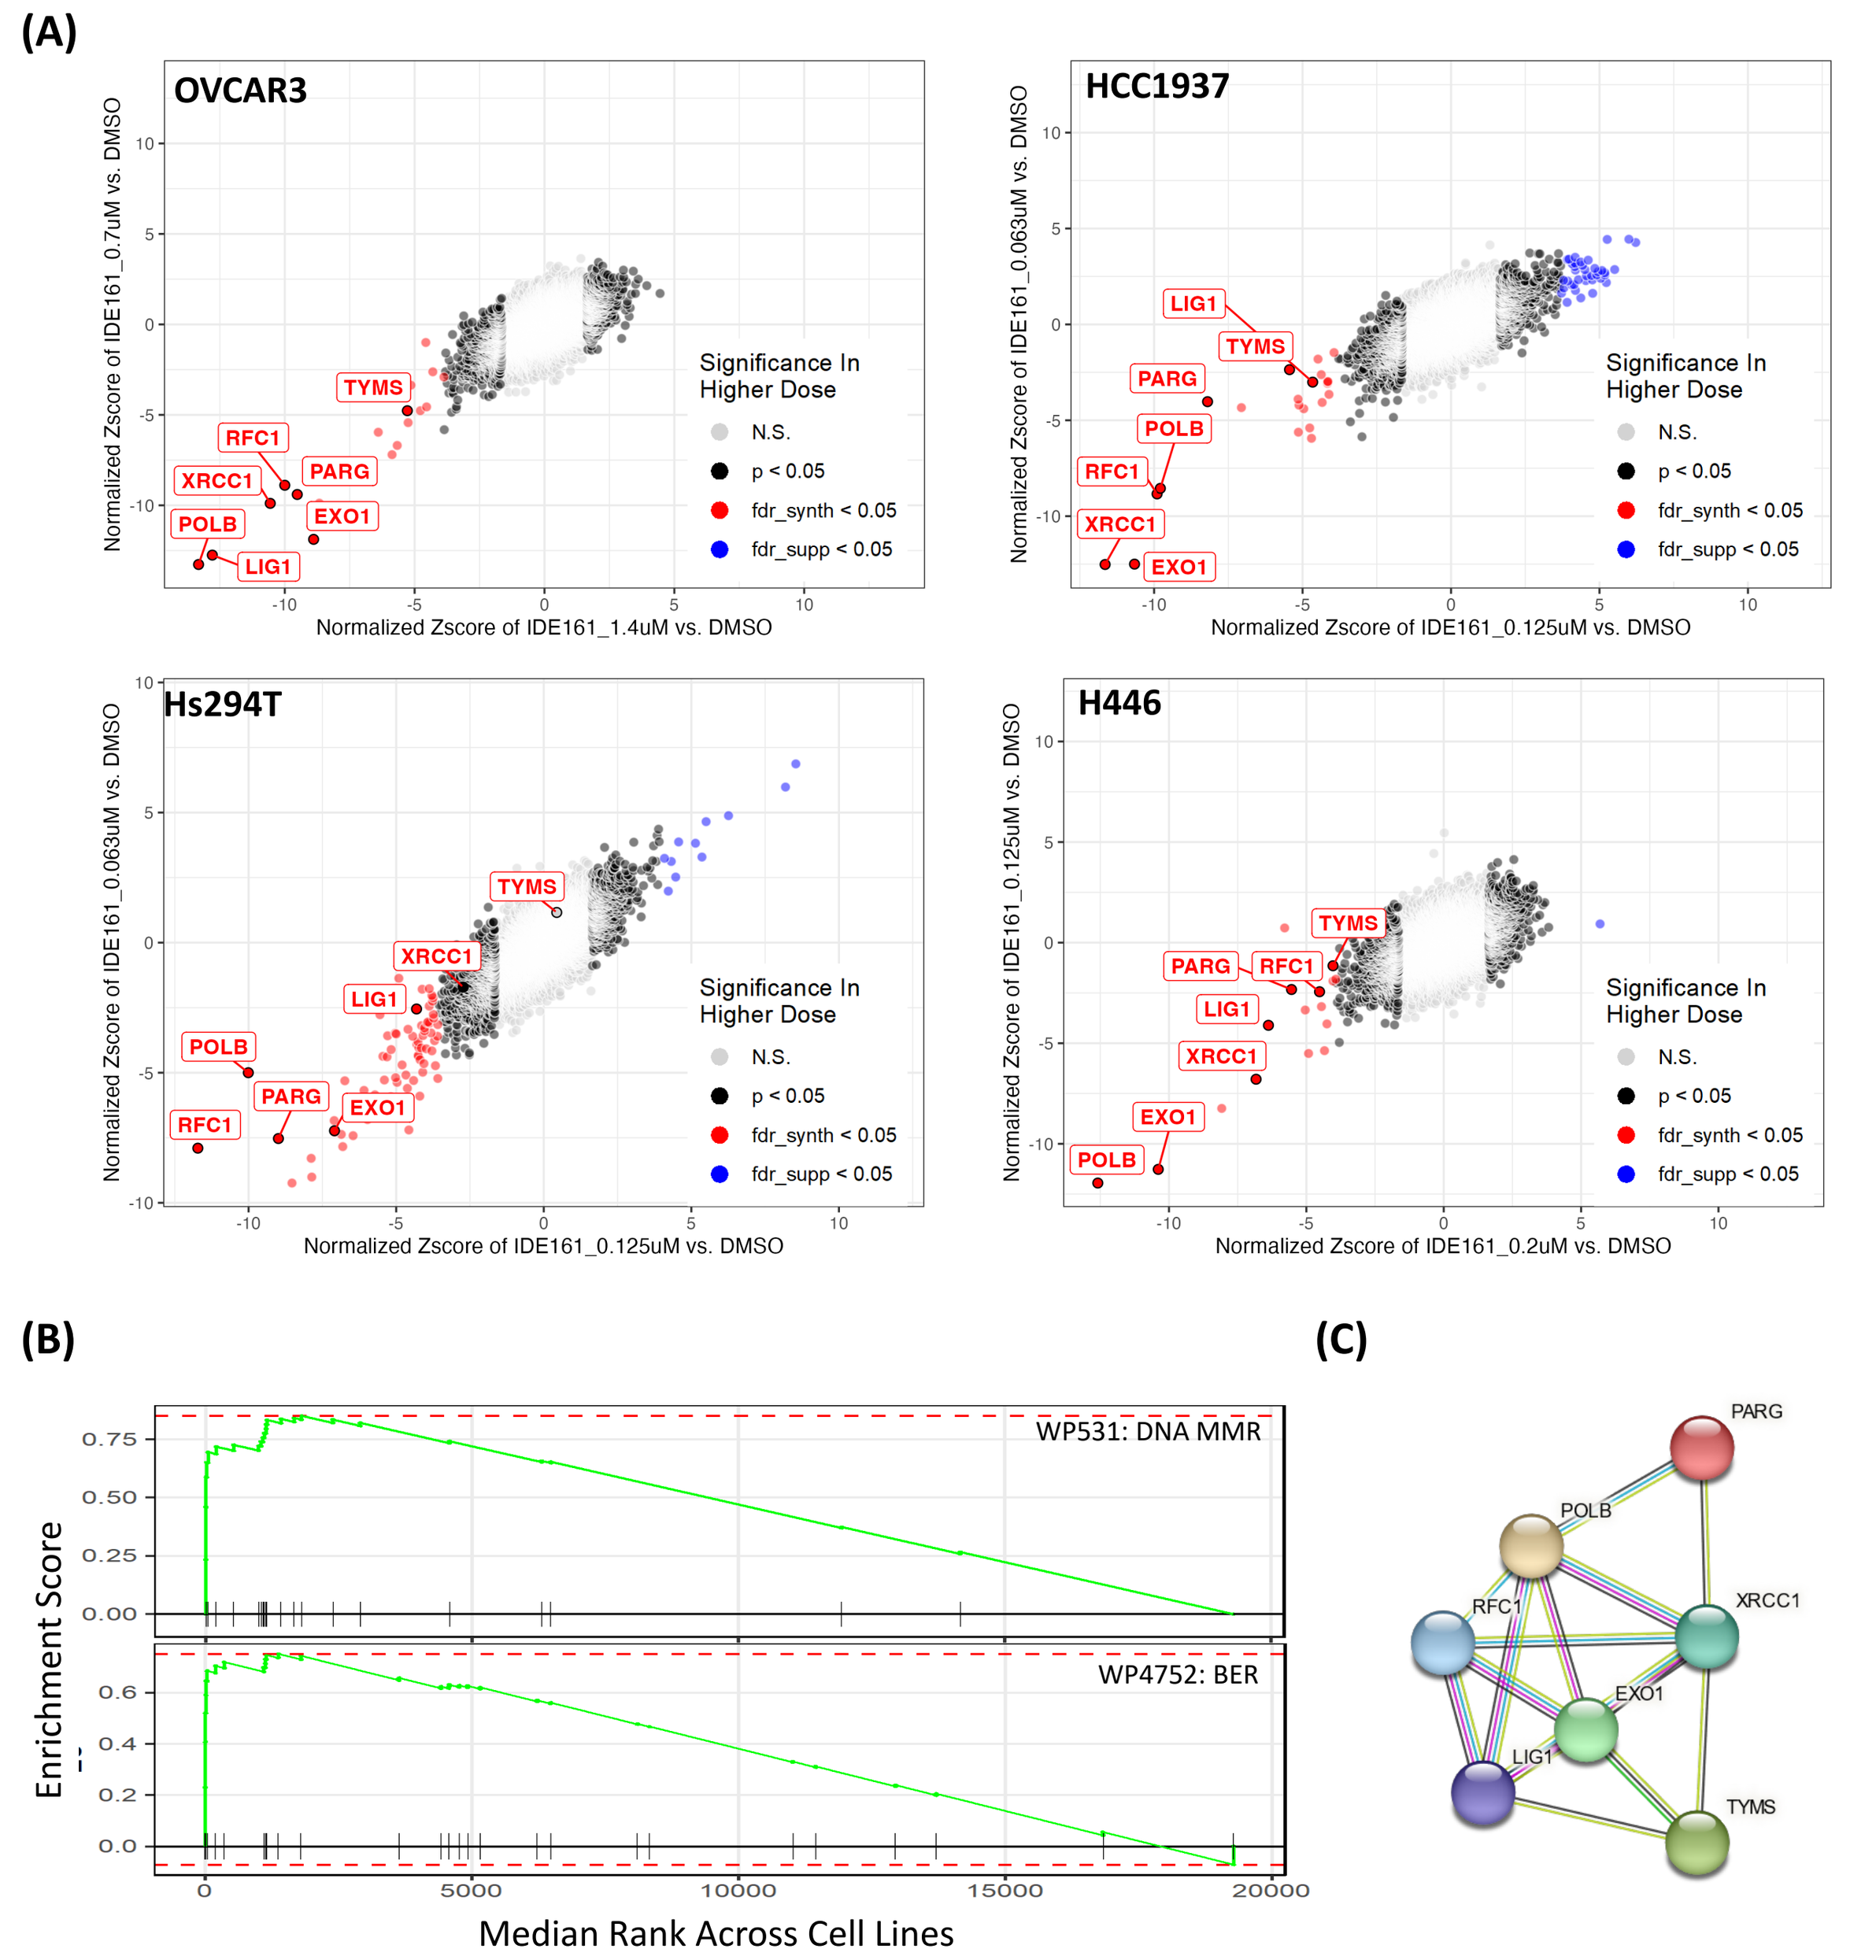
(A)
OVCAR3
HCC1937
Hs294T
H446
(B)
WP531: DNA MMR
WP4752: BER
Median Rank Across Cell Lines
(C)

(A) IDE161 shows clear synthetic lethality with DNA Damage Repair genes across tissues in whole genome CRISPR screens anchored with IDE161. Analyses conducted using drugZ. Recurrent synthetic "hits", with FDR<0.05 in 3/4 models, are labeled in all panels in red. (B) Geneset Enrichment Analysis on Median collapsed Synthetic Lethality Ranks across tissues shows strong enrichment for Base Excision Repair (BER) and DNA Mismatch Repair (MMR) pathways. Genesets obtained from WikiPathways. (C) A Gene network generated using significant Synthetic Lethal hits in at least 3 samples. Network constructed using stringDB.

- IDE161 shows synthetic lethality with DNA damage and repair genes, in particular, perturbations within the Base Excision Repair and DNA Mismatch Repair pathways
- IDE161 shows conserved synthetic lethal interactions in multiple tumor models

Figure 3: IDE161 demonstrates on-target cellular responses associated with HRD status

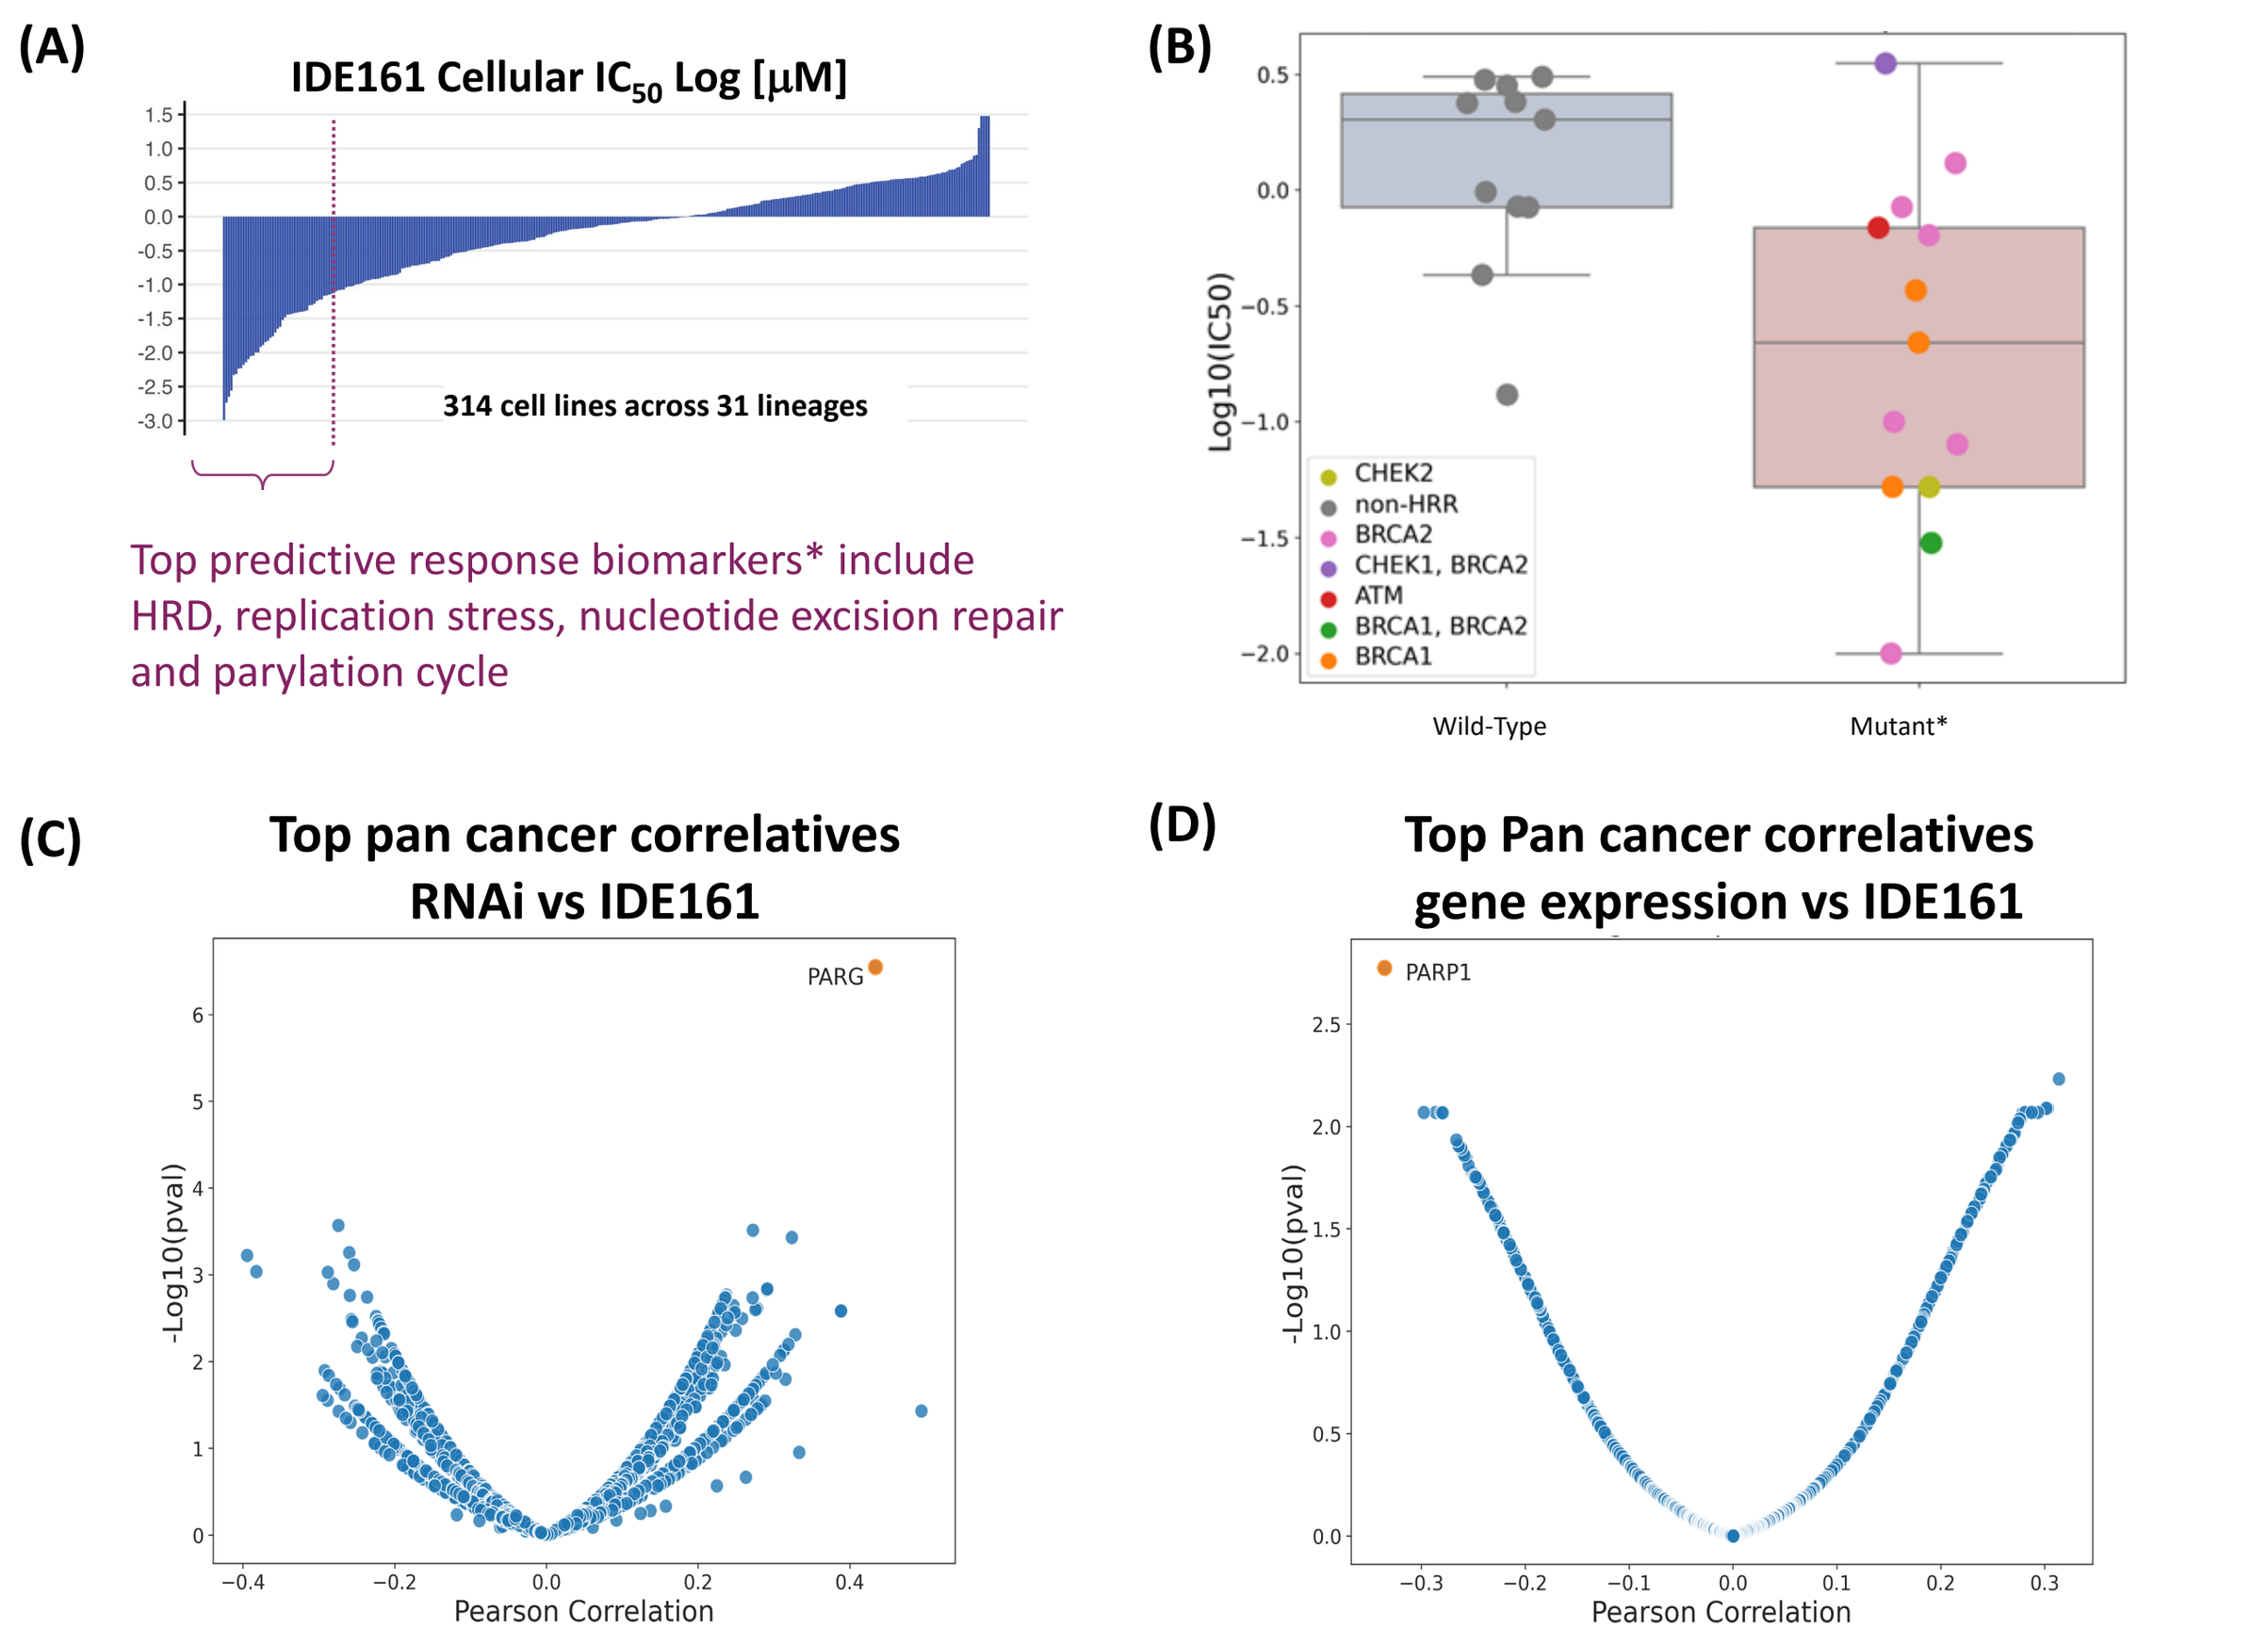
(A) IDE161 Cellular IC_{50} Log [µM]
314 cell lines across 31 lineages
Top predictive response biomarkers* include HRD, replication stress, nucleotide excision repair and parylation cycle
(B)
(C) Top pan cancer correlatives RNAi vs IDE161
(D) Top Pan cancer correlatives gene expression vs IDE161

(A) Waterfall plot of IDE161 IC_{50} values of 314 cell lines across 31 lineages representing a diversity of indications. (B) Boxplot showing IDE161 response vs the HR status of Breast Cancer cell lines (based on damaging mutants in the labeled genes). (C, D) Volcano plot showing univariate correlation between IDE161 response and shRNA Gene Effect, and RNASeq Gene Expression respectively. Cell line genetic data obtained from DepMap.

- The effect of IDE161 on the proliferation of a panel of 314 cancer cell lines across 31 lineages revealed mechanistic associations with PARGi sensitivity
- Cellular antiproliferative responses to IDE161 are strongly associated with HRD status in breast cancer cell lines
- Top pan-cancer molecular associations with IDE161 sensitivity indicate on-target cellular activity

Figure 4: Acquired PARPi resistant models support HRD as a biomarker indicating potential context-dependent rationale for PARGi post PARPi

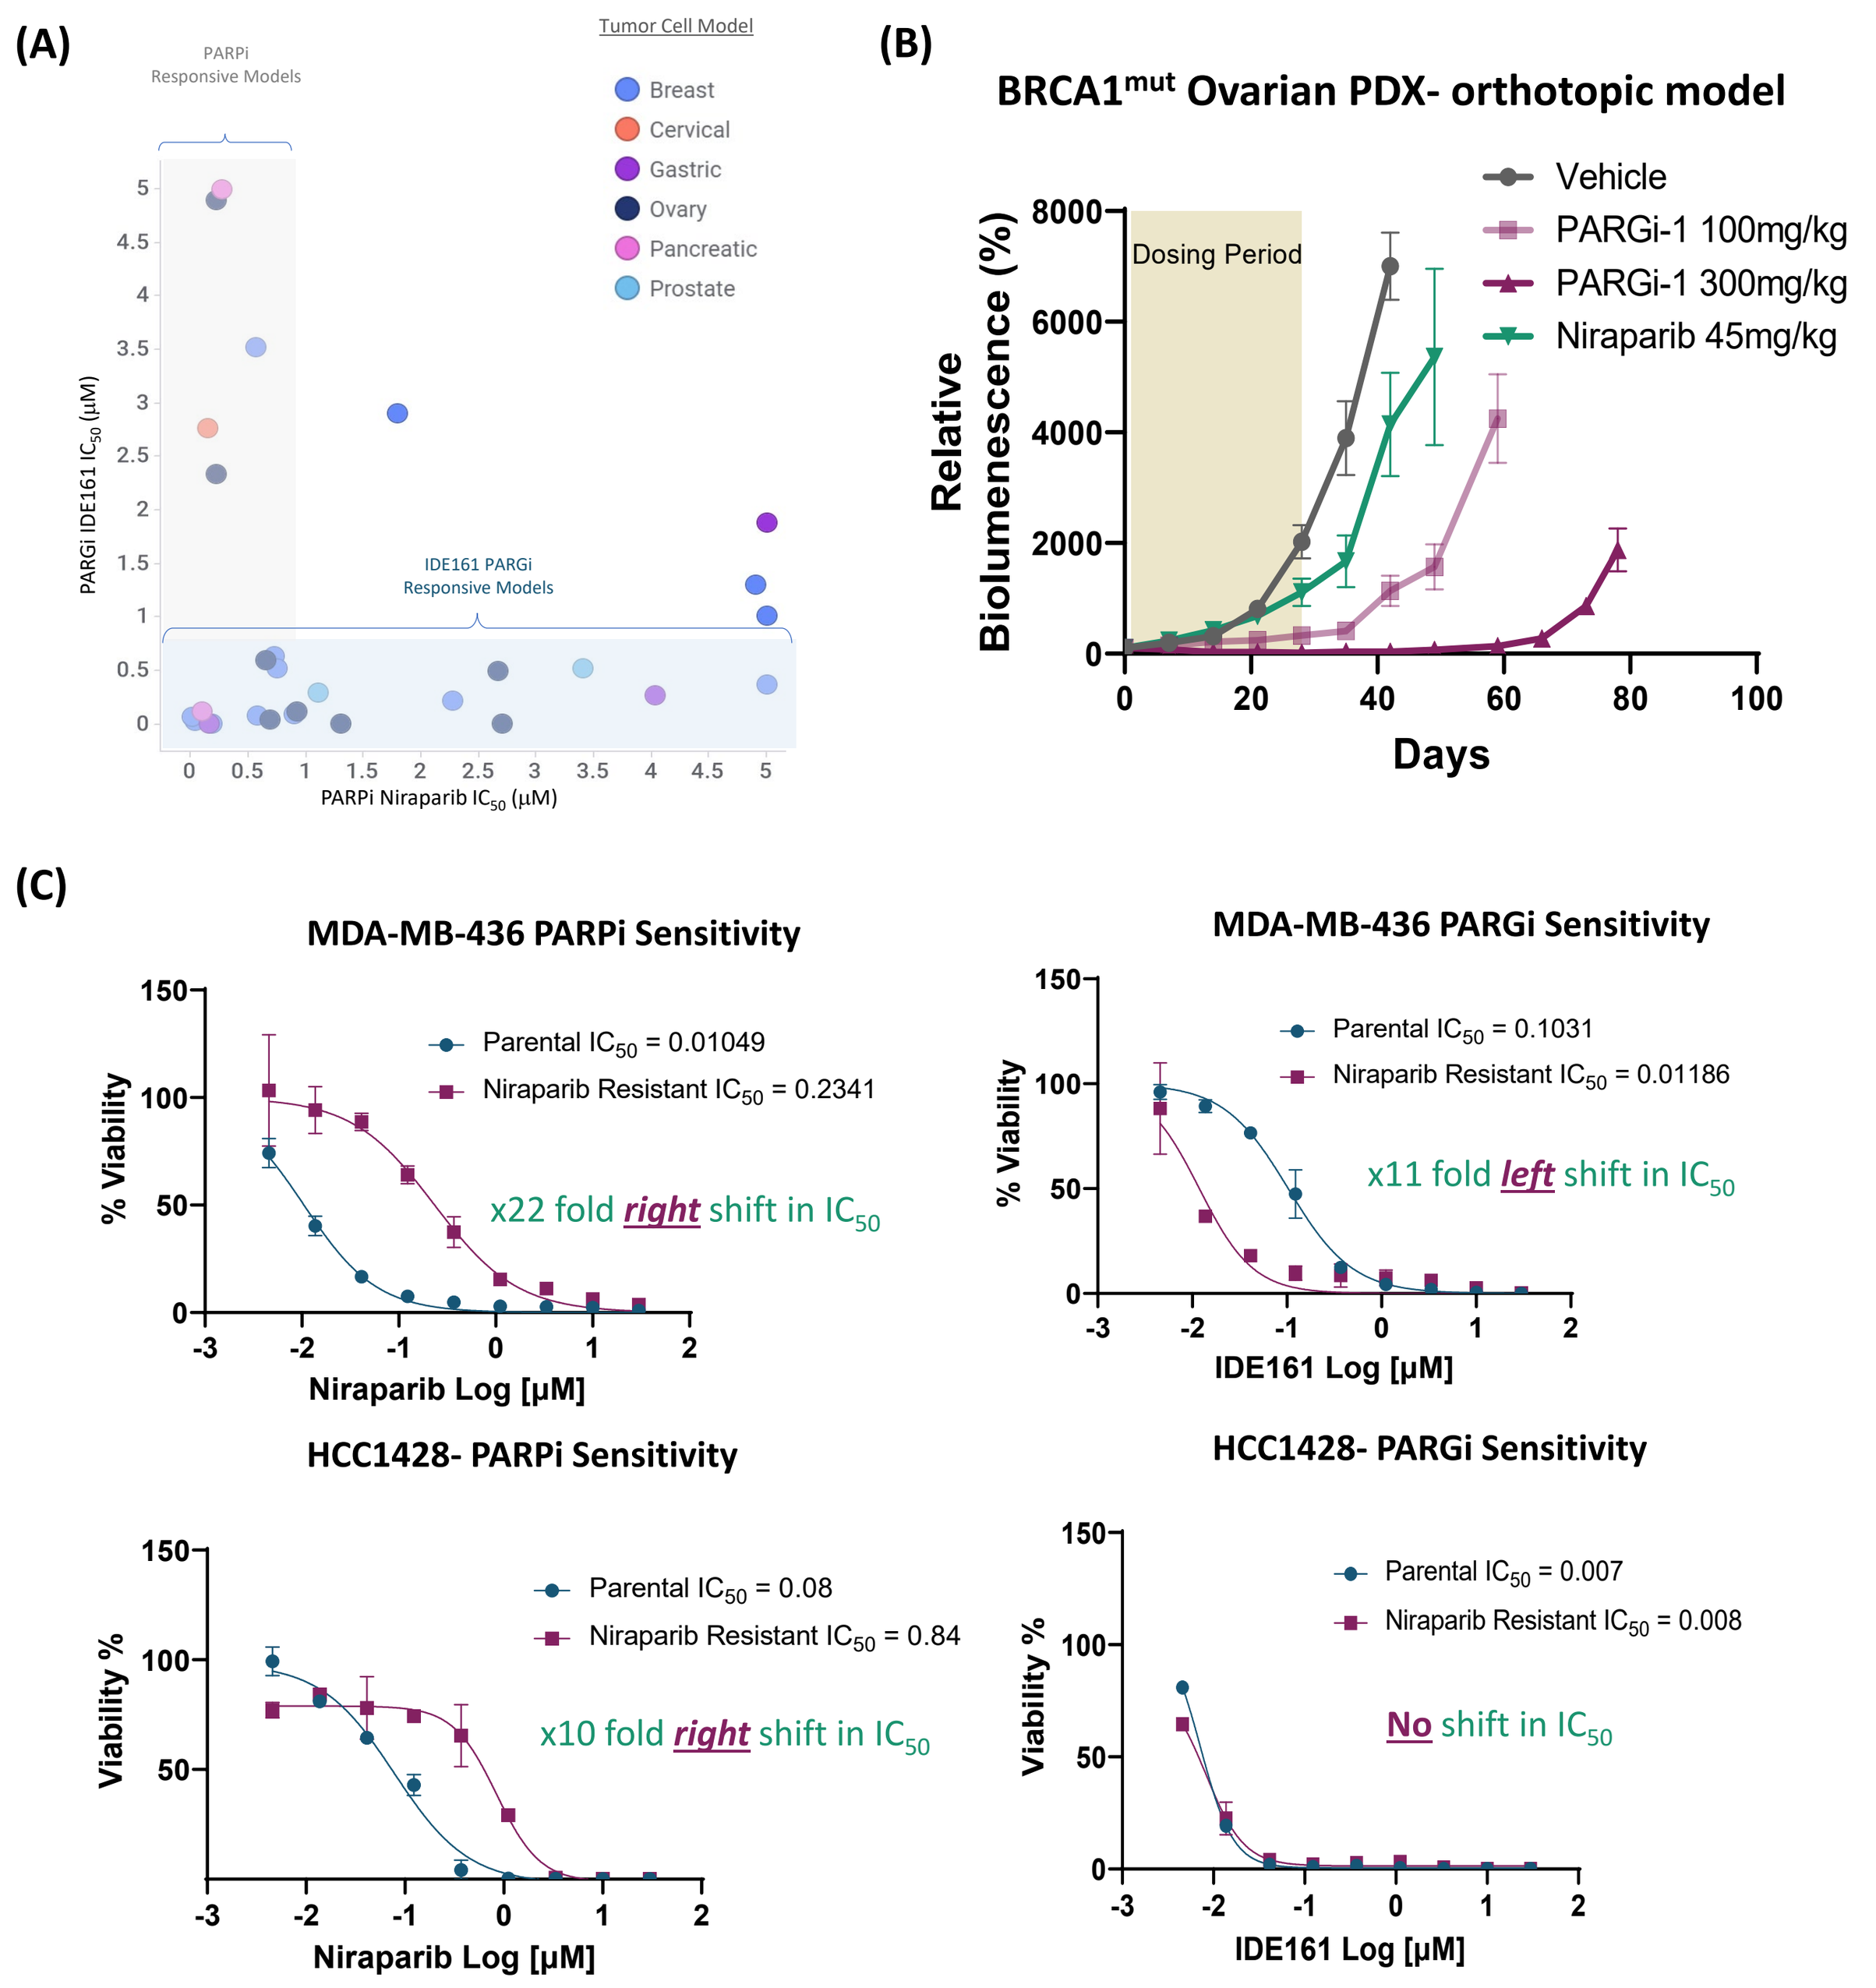
(A)
PARPi Responsive Models
IDE161 PARGi Responsive Models
(B) $BRCA1^{mut}$ Ovarian PDX- orthotopic model
Dosing Period
(C)
MDA-MB-436 PARPi Sensitivity
x22 fold right shift in IC_{50}
MDA-MB-436 PARGi Sensitivity
x11 fold left shift in IC_{50}
HCC1428- PARPi Sensitivity
x10 fold right shift in IC_{50}
HCC1428- PARGi Sensitivity
No shift in IC_{50}

(A) The effect of nirparib and IDE161 was evaluated in a panel of indicated HRD cell lines (B) NSG mice bearing luciferized PDXs were treated daily with 100 and 300 mg/kg PARGi-1 and daily with 45 mg/kg Niraparib for 28 days. After this time, mice were monitored for another 50 days to determine tumor re-growth off treatment. Tumor growth was measured weekly through bioluminescence imaging and plotted as mean ± SEM. (C) Breast cancer cells with acquired resistance to Niraparib were tested with IDE161 in a cellular viability assay. Values are plotted as mean ± SD.

- IDE161 shows selective sensitivity in HRD cell lines and differentiation from PARPi across distinct indications
- Evaluation of clinically derived PARPi resistant PDXs with PARGi-1 demonstrate durable response leading to regressions in 1/4 models tested
- Breast cancer cells with acquired resistance to Niraparib continue to be sensitive or become more sensitive to IDE161

Figure 5: PARG inhibition results in robust TGI in HRD cell (CDX) and patient-derived xenografts (PDX) with activity distinct from PARPi

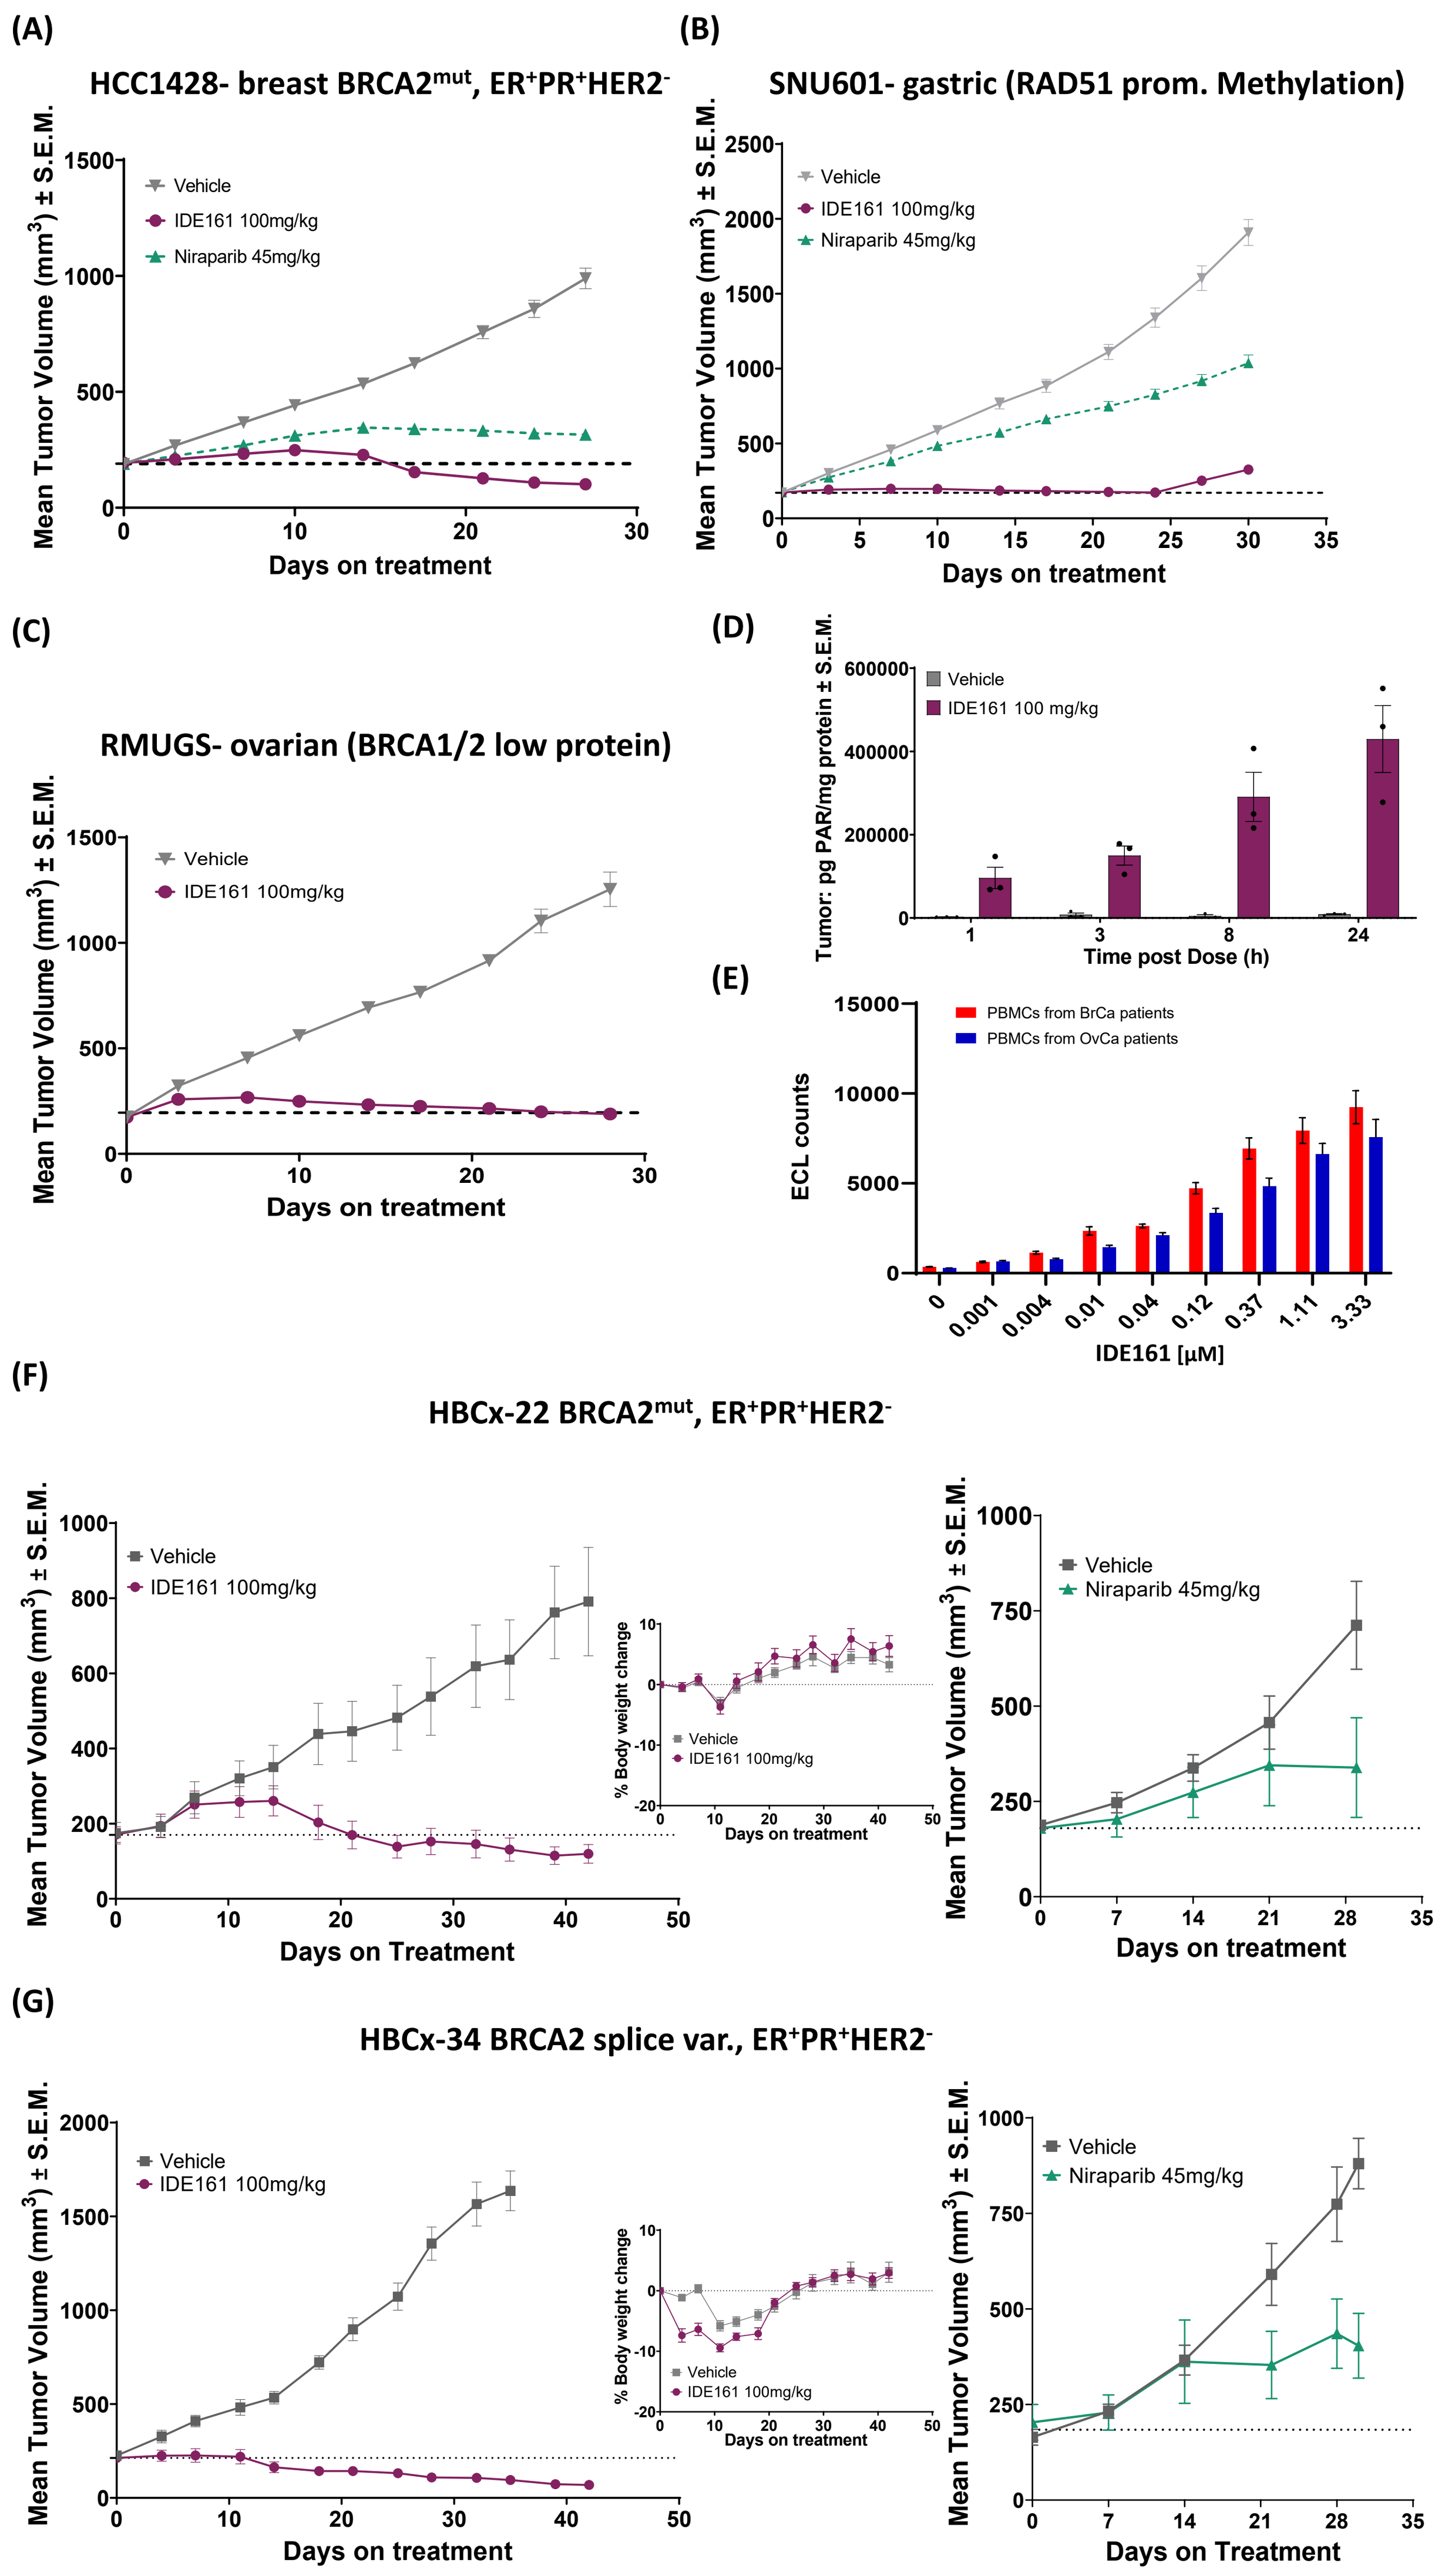
(A) HCC1428- breast $BRCA2^{mut}$, $ER^+PR^+HER2^-$
(B) SNU601- gastric (RAD51 prom. Methylation)
(C) RMUGS- ovarian (BRCA1/2 low protein)
(D)
(E)
(F) HBCx-22 $BRCA2^{mut}$, $ER^+PR^+HER2^-$
(G) HBCx-34 BRCA2 splice var., $ER^+PR^+HER2^-$

(A-C)) HRD breast, gastric and ovarian cell-derived xenograft models (CDX) were treated with 100mg/kg IDE161 or 45mg/kg Niraparib once daily, orally. Tumor measurements are represented as mean ± SEM. (D-E) PAR chain accumulation was evaluated using a PAR MSD assay in (D) ovarian CDX tumor samples and (E)) ex vivo treated PBMCs. (F-G) HRD Breast cancer patient derived xenografts (PDX) were treated with 100mg/kg of IDE161 or Niraparib at 45mg/kg once daily, orally- in two independent studies. Tumor measurements are represented as mean ± SEM

Dashed line; Mean tumor volume D0

- Tumor growth stasis and regressions observed in HRD breast, gastric and ovarian cell-derived xenograft models (CDX).
- Breast cancer patient-derived xenografts (PDX) treated with IDE161 demonstrate durable regressions as compared to the tumor stasis achieved with PARPi in the same models; highlighting a differentiated therapeutic MOA between PARG and PARP inhibitors.
- Poly-PAR chain accumulation is a robust and quantitative PD response to IDE161 in tumor samples and ex vivo treated human PBMCs supporting its use as a peripheral biomarker to assess exposure/response relationships.

Acknowledgments

- PAR ELISA studies were performed by Quintara
- CRISPR anchor studies were performed by Cellecta, Inc.
- in vivo CDX studies were performed at Pharmaron
- in vivo PDX studies were performed at Xentech and DFCI